Update on Phase 1/2 study of VAL-083 (dianhydrogalactitol) in patients with recurrent malignant glioma

Kent C. Shih[1,2], Manish Patel[1,3], Nicholas Butowski[4], Sani Kizilbash[5], Jeffrey Bacha[6], Dennis Brown[6], Anne Steino[6], Richard Schwartz[6], Sarath Kanekal[6], Lorena Lopez[6], and Howard A. Burris, III[1,2]* *[1]Sarah Cannon Research Institute, Nashville, USA; [2]Tennessee Oncology, Nashville, USA; [3]Florida Cancer Specialists & Research Institute, Florida, USA; [4]University of California San Francisco Division of NeuroOncology, [5]Mayo Clinic, Rochester, USA; [6]DelMar Pharmaceuticals, Inc., Vancouver, Canada and California, USA*

ABSTRACT # 481

(ClinicalTrials.gov Identifier: NCT01478178):

Poor GBM outcomes correlate with high MGMT expression. VAL-083 is a bi-functional alkylating agent causing DNA cross-links at N7 of guanine. Clinical activity against GBM was shown in prior NCI-sponsored trials. We have shown VAL-083 cytotoxic activity against GBM cell lines independent of MGMT. This clinical trial determined an optimized dose regimen to advance VAL-083 into Phase 2/3 trials for refractory GBM. Patients received a single dose of VAL-083 on days 1 - 3 of a 21-day cycle, v. days 1 – 5 in a 35 day cycle in prior NCI trials. 30 patients with histologically-confirmed GBM were enrolled in 8 dose cohorts (1.5 to 50mg/m²/d) in an open-label, single-arm Phase 1/2 dose-escalation study (3+3 design). All patients had failed prior therapy including temozolomide and bevacizumab, unless contraindicated. Preliminary analysis suggested a dose-dependent survival benefit: median OS=9.0 months (≥30mg/m²/d) v. OS=4.4 months (<10mg/m²/d). The optimized regimen achieved a higher single dose and dose intensity v. prior NCI trials without evidence of drug-related SAEs at doses ≤40mg/m²/d. DLTs (thrombocytopenia) were observed at 50mg/m²/d with platelet nadir occurring around day 20, with rapid and spontaneous recovery consistent with prior NCI trials. PK analyses showed dose-dependent linear systemic exposure and calculated CNS concentrations at 40mg/m²/d within the observed IC_{50} range for GBM cell-lines *in vitro*. An expansion cohort has been initiated at 40mg/m²/d. CONCLUSIONS: Our optimized VAL-083 dose regimen exhibited favorable safety profile at 40mg/m²/d with a trend toward improved survival v. lower doses. Additional data from this ongoing clinical trial will be presented.

Table 1:
Characteristics of Patients Receiving VAL-083

	DOSE OF VAL-083	
	<10mg/m2 ***(n=10)***	**≥30mg/m2** ***(n=6)***
Age: Mean (range)	49 (30-38	61 (23-71)
Sex: M / F	3 / 7	4 / 1
KPS: Mean (range)	80 (60-100)	85 (60-100)
Initial lesion		
R temporal	2	2
L temporal	2	0
R parietal	1	2
L parietal	3	0
R frontal	1	1
L frontal	0	2
L ventricular	0	0
R occipital	0	0
Not reported	1	0
Recurrence		
R temporal	2	2
L temporal	1	0
R parietal	1	1
L parietal	2	0
R frontal	1	2
L frontal	1	0
L ventricular	0	1
R occipital	0	1
L cerebellum	1	0
not reported	1	1
Prior Therapy		
temozolomide	10	6
bevacizumab	9	6
Salvage Therapy Prior to VAL-083		
temozolomide re-challenge	3	1
Irenotecan	3	0
PDGFR Antibody	2	0
Virinostat	1	0
bevacizumab re-challenge	2	2
Radiation re-challenge	1	0
carboplatin	1	1
Onartuzumab	1	0
Gleevec	1	0
BKM-120 (combination with bev)	0	1
gliadel	0	1
CCNU	0	2

RESULTS AND OBSERVATIONS

Glioblastoma multiforme (GBM) is the most common primary brain tumor in adults; despite standard therapy with surgery, radiation and chemotherapy with temozolomide, median survival is only 14.6 months. Essentially all patients develop recurrent or progressive disease after initial therapy, after which the median survival is less than six months. [1, 2]

Bevacizumab is approved as a single agent for patients with recurrent GBM following prior therapy as an alternative to corticosteroids to relieve disease symptoms; however, there are no data demonstrating an improvement in disease-related symptoms or increased survival in refractory GBM with bevacizumab.[3]

Currently there are no approved treatments for GBM patients failing bevacizumab and rapid clinical deterioration is often observed after discontinuing bevacizumab.[4] For patients receiving supportive care, median survival is only 2 months while a range of salvage chemotherapies demonstrate median survival of 5.2 months after bevacizumab failure.[5]

VAL-083 is a bi-functional alkylating agent that readily crosses the blood brain barrier.[6] VAL-083 demonstrated activity against a range of tumor types, including GBM, in prior clinical trials sponsored by the US National Cancer Institutes (NCI). The cytotoxic mechanism of VAL-083 has been shown to be independent of the MGMT-repair mechanism suggesting the potential to treat temozolomide-refractory GBM patients.

We recently completed a dose-escalation study with VAL-083 in GBM patients whose tumors had progressed following standard treatment with temozolomide, radiotherapy, bevacizumab and a range of salvage therapies. No drug-related severe adverse events were reported and myelosuppression was mild at doses ≤40mg/m²/day. Dose limiting toxicity was observed at a dose of 50mg/m²/day (See Table 2).

Preliminary data analysis suggested a dose-dependent and clinically meaningful survival benefit in this population.

Further sub-group analysis for patients receiving up to 5 mg/m² daily x 3 every 21 days (low dose) versus those patients receiving 30 mg/m² or 40 mg/m² (high dose) serve to diminish the possibility that factors other than the proposed clinical activity of VAL-083 are responsible for the observed dose-dependent survival benefit following treatment.

- **Following initiation of VAL-083 treatment, patients in the low dose sub-group had a median survival of 5.1 months versus median survival of 9.2 months for patients in the high dose sub-group (p=0.04).** ***(See Figure 1 and Table 3)*****.**
- **Increased survival was observed at 6, 9 and 12 months following initiation of treatment in the high-dose sub-group compared to the low dose sub-group** ***(See Table 3)*****.**
- **Survival was not correlated with screening Karnofsky performance status (R^2_{KPS} = 0.03) or subject age at screening (R^2_{AGE} = 0.01)**
- **Survival was not correlated with the localization of either the initial or recurrent lesion**
- **A longer time to progression on front-line temozolomide or second line bevacizumab treatment was not correlated with a longer survival benefit following initiation of VAL-083 therapy.**
- **All patients had failed one or more salvage therapies prior to initiation of VAL-083. No commonalities in prior therapy were observed to correlate with survival** ***(See Table 1)*****.**
- **Survival was not correlated with MGMT expression in either the low dose or high dose sub-group**

MGMT Analysis

For patients whose data or tissue blocks were available for analysis, MGMT expression was evaluated. Methylation status of the *MGMT* promotor was characterized by PCR by the treating physician for eight patients enrolled to date in the Phase I dose-escalation and Phase II expansion phase. Seven (87.5%) were reported as unmethylated, thus expected to be high expressers of MGMT and resistant to temozolomide-based treatment regimens.

For 13 of the patients, slide-fixed tissue was available for ELISA detection of MGMT protein. The results showed 4/13 (31%) patients were expressing lower levels of MGMT (32-41 ng MGMT/mg total protein), while 9/13 (69%) patients expressed higher levels of MGMT (67-182 ng MGMT/mg total protein).

In line with previous data from other studies using different methods of MGMT detection, these results show that the majority of GBM patients have unmethylated *MGMT* promoter regions and thus express high levels of MGMT, which make them unlikely to respond to TMZ.[7,8]

In the low VAL-083 dose sub-group (<10mg/m²), two of ten patients (20%) have known MGMT status. Both are unmethylated (high expressers of the MGMT protein). In the high VAL-083 dose sub-group (≥30mg/m²) four of six patients (66%) were reported as unmethylated or high expressers of MGMT, while 2/6 (34%) were reported as methylated or low expressers of MGMT. Considering known MGMT status in both sub-groups an equal survival distribution on both sides of the median was observed suggesting that the survival outcome was *not* correlated with MGMT expression.

CONSLUSIONS AND NEXT STEPS:

- **Sub-group analysis of data from DLM-10-001 Phase I study further supports a dose-response and potential survival benefit in GBM patients failing standard front-line therapy and bevacizumab following treatment with VAL-083.**
- **A Phase 2 expansion cohort of up to 14 patients has been initiated at the well-tolerated dose of 40 mg/m²/d. Eight (8) patients have been enrolled to date.**
- **An interim 45 mg/m²/d dose is being studied in parallel with the expansion cohort. Three (3) patients have been enrolled.**
- **The expansion cohort may be continued at the 45 mg/m²/d dose if safety data warrant.**
- **Activities related to design of a proposed registration-directed Phase II/III trial have been initiated. The goal of the registration trial will be to examine the impact of extended VAL-083 treatment on OS and radiographic tumor response in patients with refractory GBM.**

Table 2. Hematologic toxicities observed in patients in DLM-10-001 trial (CTCAE Grading).

Dose		≤ 20mg/m²	30mg/m²	40mg/m²	50mg/m²
Anemia *(Hct; Hg)*	G1	-	-	1/3 *(33.3%)*	5/6 *(83.3%)*
	G2	-	-	-	-
	G3	-	-	-	-
	G4	-	-	-	-
Leukopenia *(WBC)*	G1	-	1/3 *(33.3%)*	1/3 *(33.3%)*	3/6 *(50.0%)*
	G2	-	-	-	2/6 *(33.3%)*
	G3	-	-	-	2/6 *(33.3%)*
	G4	-	-	-	-
Neutropenia *(neutrophils)*	G1	-	-	1/3 *(33.3%)*	2/6 *(33.3%)*
	G2	-	-	-	-
	G3	-	-	-	1/6 *(16.6%)*
	G4	-	-	-	1/6 *(16.6%)*
Thrombocyto-penia *(platelets)*	G1	-	2/3 *(66.6%)*	2/3 *(66.6%)*	1/6 *(16.6%)*
	G2	-	-	-	-
	G3	-	-	-	3/6 *(50.0%)**
	G4	-	-	-	1/6 *(16.6%)**

**DLT observed in 2 of 6 (33.3%) patients at 50mg/m², as defined by Grade 4 thrombocytopenia in 1 of 6 patients (16.6%) and Grade 3 thrombocytopenia with hemorrhage in 1 of 6 patients (16.6%).*

Figure 1: Survival of post-bevacizumab GBM patients following VAL-083 treatment

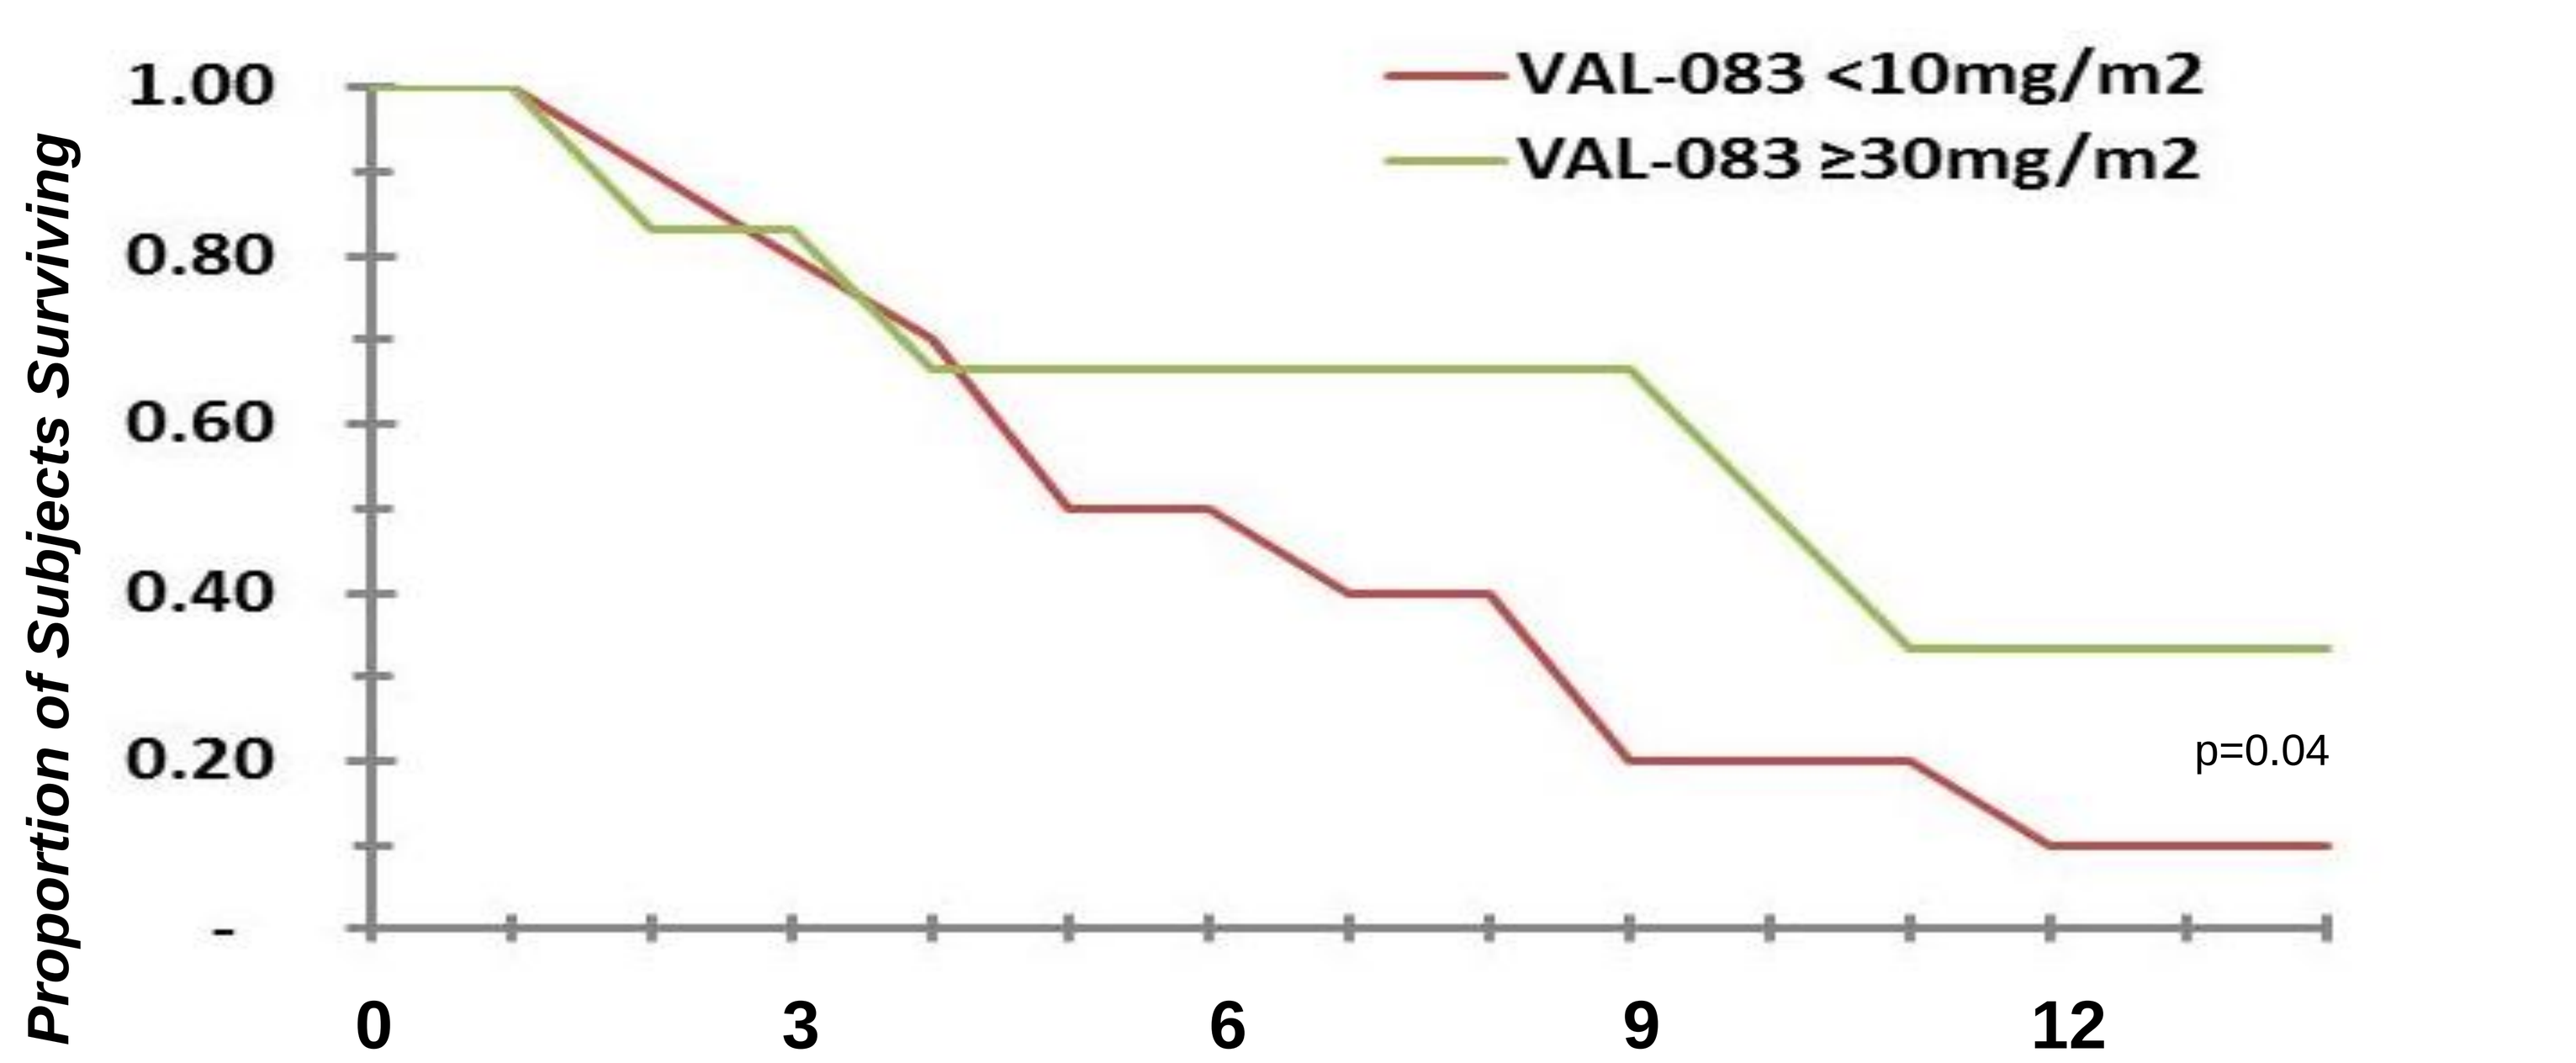
VAL-083 <10mg/m2
VAL-083 ≥30mg/m2
1.00
0.80
0.60
0.40
0.20
-
Proportion of Subjects Surviving
p=0.04
0
3
6
9
12
Months from initiation of VAL-083 therapy

Table 3: Percent & median Survival of post-bevacizumab GBM patients following VAL-083 treatment

Dose Cohort	6 months	9 months	12 months	median
High (≥30mg/m²)	67%	67%	33%	9.2 months
Low (<10mg/m²)	44%	33%	22%	5.1 months

PHARMACOKINETICS:

Pharmacokinetic analysis further supports VAL-083 culpability for the observed dose-dependent survival benefit. Calculated tumor tissue concentrations suggest that patients in the low dose sub-group are likely to have received a sub-therapeutic dose, whereas patients in the high dose sub-group should achieve tumor tissue concentrations of VAL-083 at or above the IC_{50} for multiple GBM cell lines *in vitro* (See Table 4).

Fig 2. **PK by dose cohort & published literature[9]**

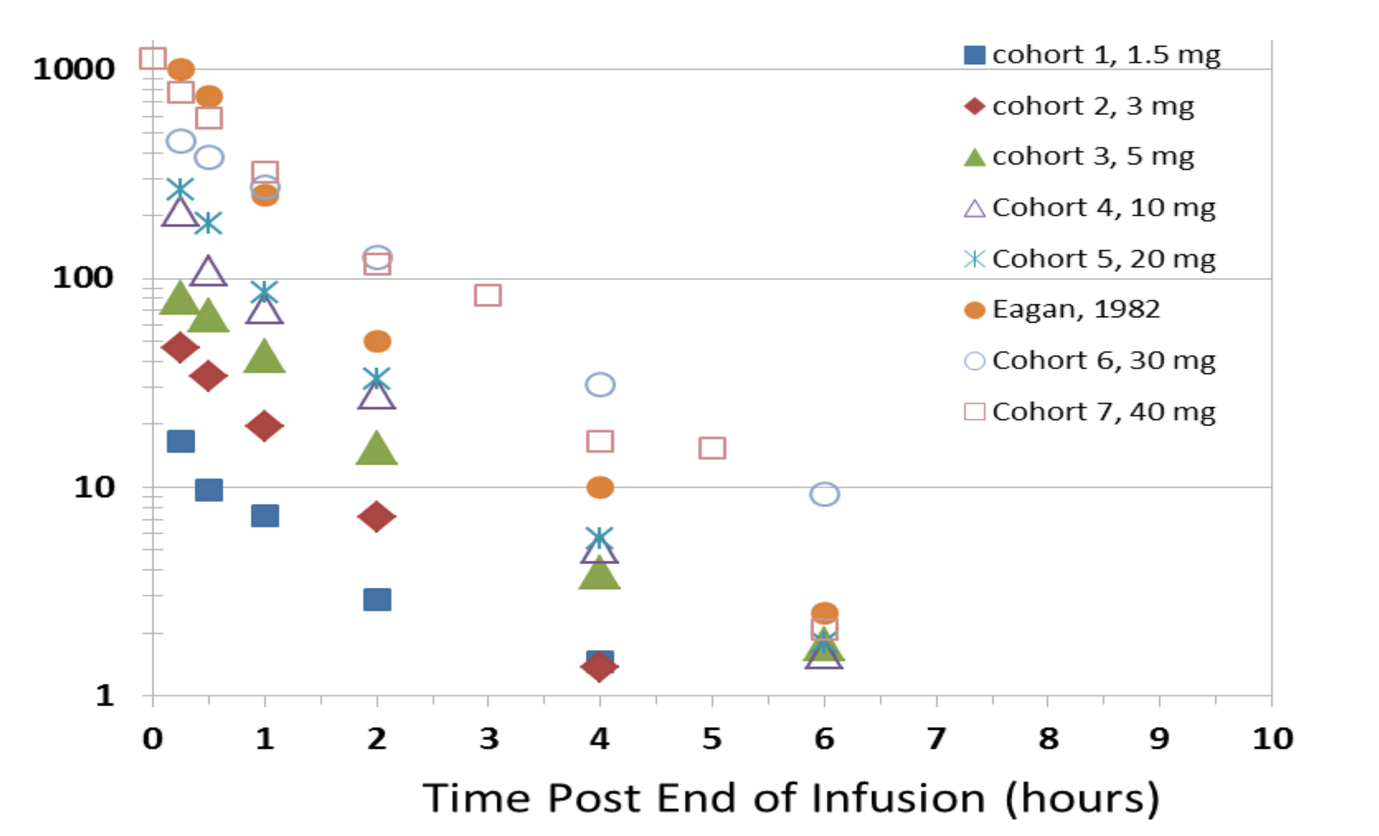
1000
100
10
1
cohort 1, 1.5 mg
cohort 2, 3 mg
cohort 3, 5 mg
Cohort 4, 10 mg
Cohort 5, 20 mg
Eagan, 1982
Cohort 6, 30 mg
Cohort 7, 40 mg
0
1
2
3
4
5
6
7
8
9
10
Time Post End of Infusion (hours)

Fig 3. **Dose-AUC relationship**

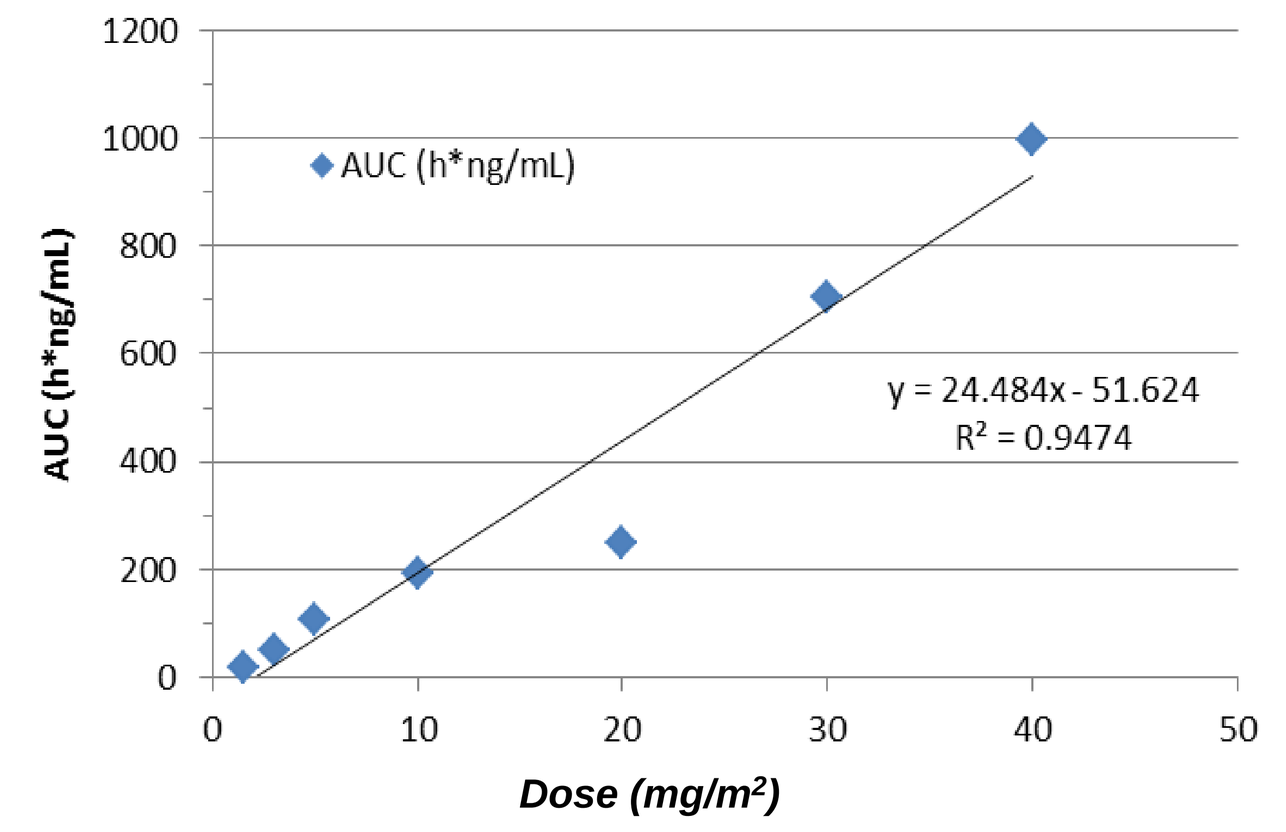
1200
1000
800
600
400
200
0
AUC (h*ng/mL)
AUC (h*ng/mL)
y = 24.484x - 51.624
R² = 0.9474
0
10
20
30
40
50
Dose (mg/m²)

Table 4. Estimated Tumor Concentration in Human Brain Exceeds in vitro IC_{50} for 40 mg/m² dose

Dose x 3 days	Plasma Cmax (µg/mL)[a]	Estimated Maximum Tumor Concentration in Brain [b,c]		IC_{50} in GBM Cell Lines**
		(µg/g tissue)	µM*	µM
5mg/m²	0.081	0.059	0.40	2 – 4
40mg/m²	0.781	0.563	3.86	

a. From study DLM-10-001, cohort #7; PK was conducted only on Day 1, given the short t-1/2 of ~1h Cmax is assumed to be same for Day 2 & 3.
b. Percent of plasma drug concentration in brain tumor = 44%, Eckhardt, 1977
c. Half-life of drug in human brain tumor tissue = 20h, Eckhardt, 1977
*Volume of 1 g tissue assumed to be 1 mL
**IC_{50} range for low MGMT (U251 and SF188) and high MGMT (T98G) GBM cells tested

References:

1. Wong et al. Outcomes and prognostic factors in recurrent glioma patients enrolled onto phase II clinical trials J Clin Oncol 1999;17:2572-2578
2. Lamborn et al. Progression-free survival: an important end point en evaluating therapy for recurrent high-grade glioma. Neuro Oncol 2008; 10:162-170
3. As stated in Avastin (Bevacizumab) prescribing Information: http://www.gene.com/download/pdf/avastin_prescribing.pdf
4. Anathnarayan et al. Time course of imaging changes of GBM during extended treatment. J. Neurooncol 2008;88:339-347
5. Iwamoto et al. Patterns of relapse and prognosis after bevacizumab failure in recurrent glioblastma. Neurology 2009; 73:1200-1206
6. Eckhardt et al. Cancer Treat Rep. 1977; 61(5):841-7
7. Hegi et al. MGMT gene silencing and benefit from temozolomide in glioblastoma. N Engl J Med. 2005 Mar 10;352(10):997-1003.
8. Christians et al. Prognostic value of three different methods of MGMT promoter methylation analysis in a prospective trial on newly diagnosed glioblastoma. PLoS One. 2012;7(3):e33449
9. Eagan R.T. et al. Cancer Treat Rep. 1982; 66(8):1647-49.